ABSTRACT/ POSTER 35076

TP53 WILDTYPE STATUS CAN PREDICT SENSITIVITY TO XPO1 INHIBITORS IN PATIENT-DERIVED CANCER MODELS

M. E. Maloof[1], A. A. Ellero[1], P. Taverna[1], C.J. Walker[1]

[1]Karyopharm Therapeutics Inc., Newton, MA

INTRODUCTION

- Selinexor (SEL) is a selective inhibitor of exportin 1 (XPO1) that leads to the nuclear accumulation and activation of key tumor suppressors (e.g., p53, p21, RB, and FOXOs), resulting in selective cancer cell death (Figure 1). Eltanexor (ELT) is an investigational compound with the same target and similar properties.

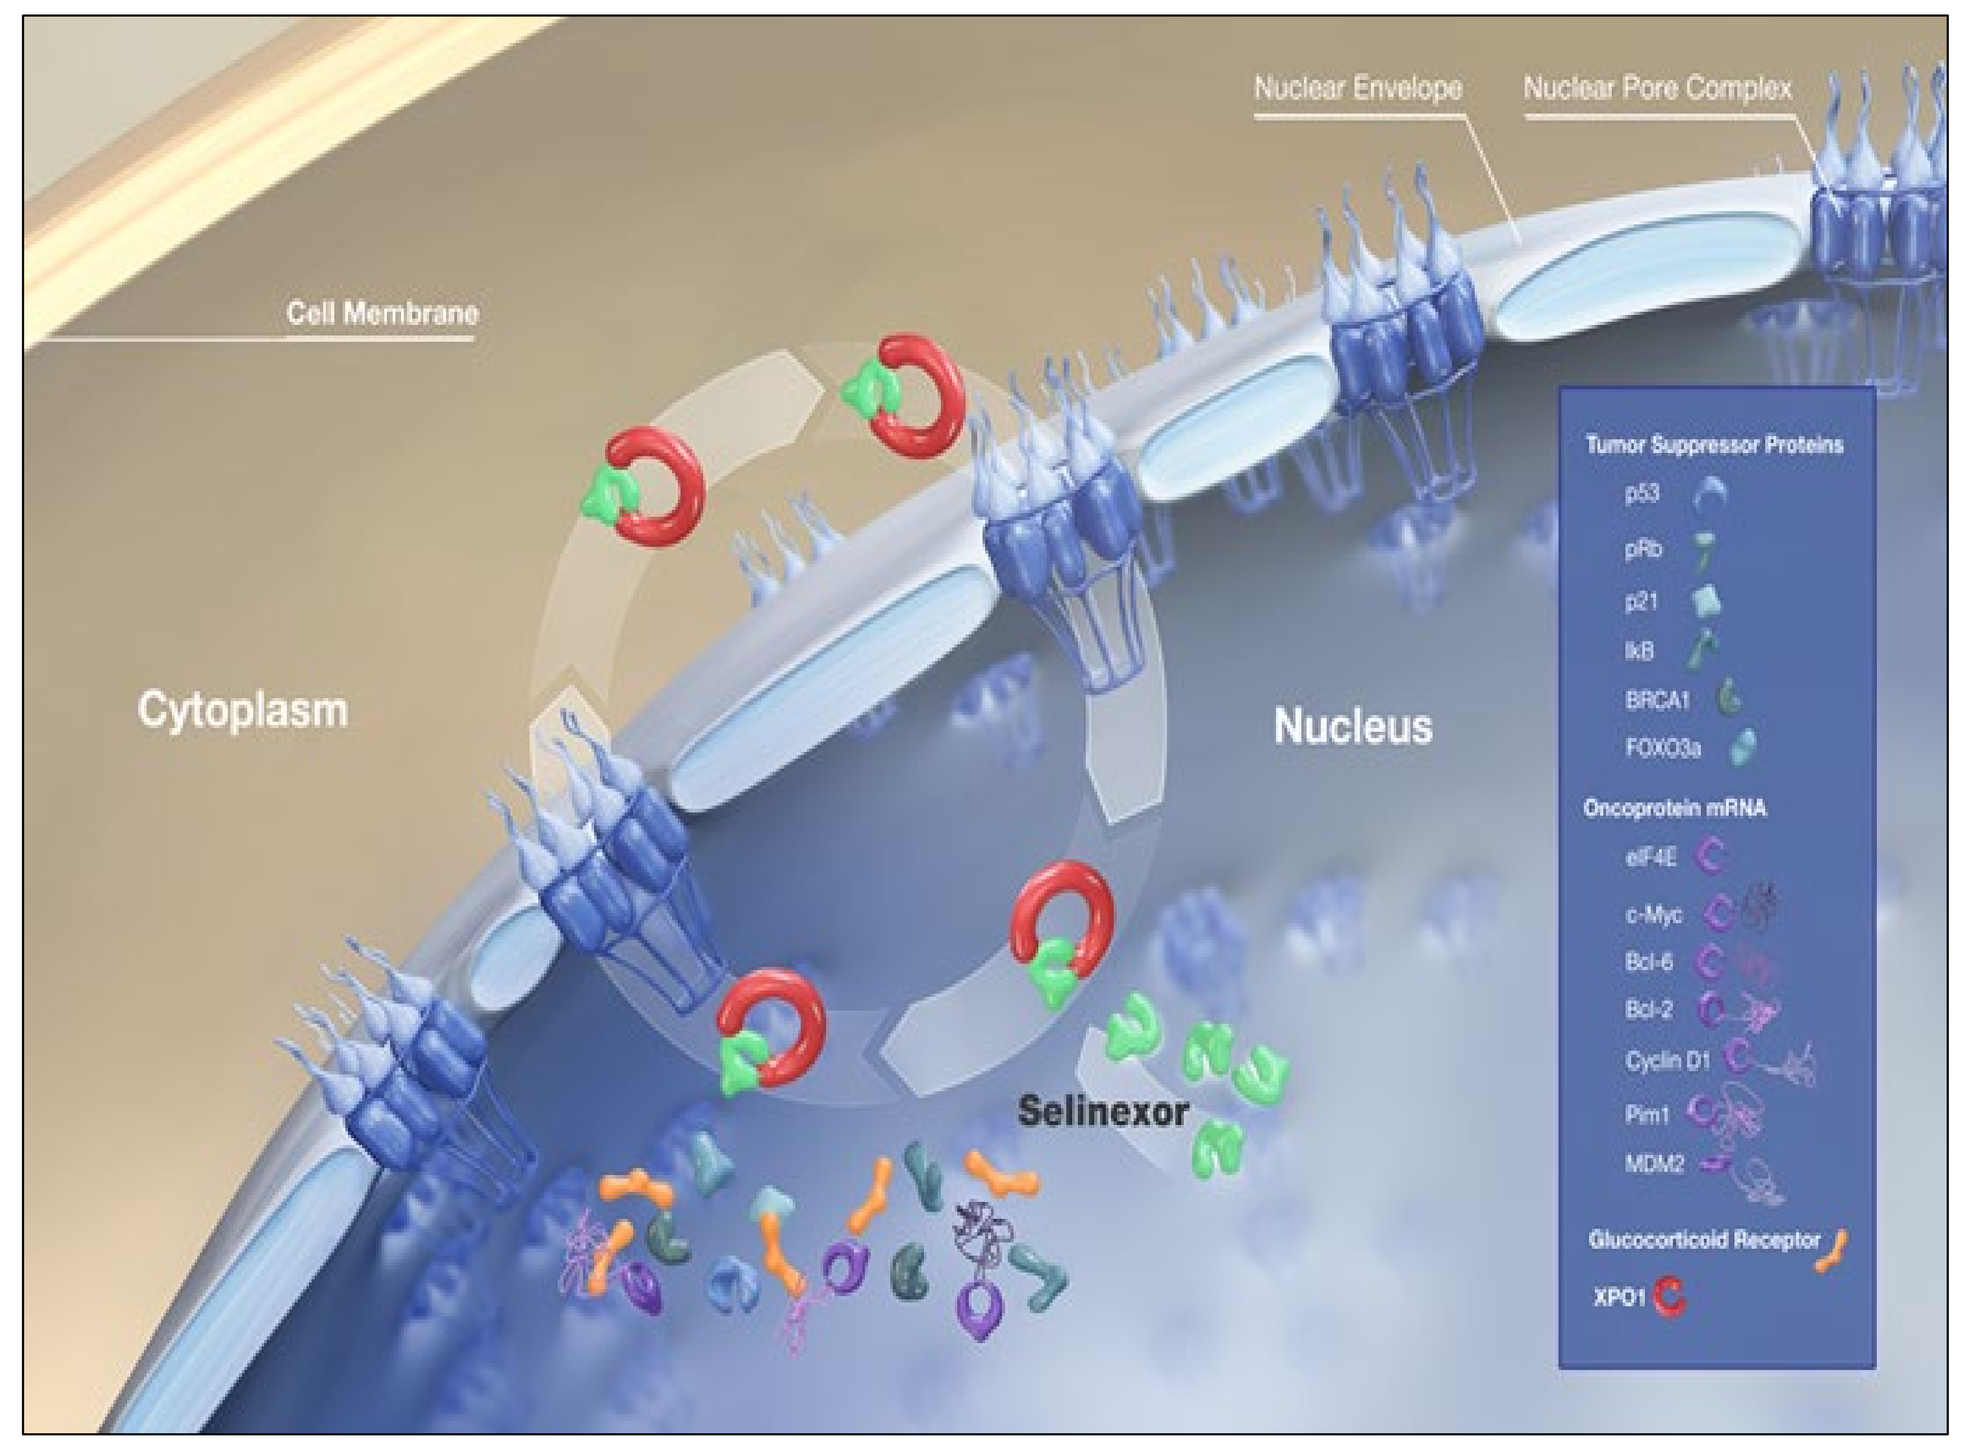
Nuclear Envelope
Nuclear Pore Complex
Cell Membrane
Cytoplasm
Nucleus
Selinexor
Tumor Suppressor Proteins
p53
pRb
p21
IkB
BRCA1
FOXO3a
Oncoprotein mRNA
eIF4E
c-Myc
Bcl-6
Bcl-2
Cyclin D1
Pim1
MDM2
Glucocorticoid Receptor
XPO1

Figure 1. Selinexor mechanism of action

- In the exploratory analysis of the SIENDO trial evaluating SEL as a maintenance therapy for patients with advanced stage or relapsed endometrial cancer (EC), longer PFS was seen in the SEL arm compared to placebo for the subset of patients with *TP53* wild-type (WT) EC (Figure 2).[1]

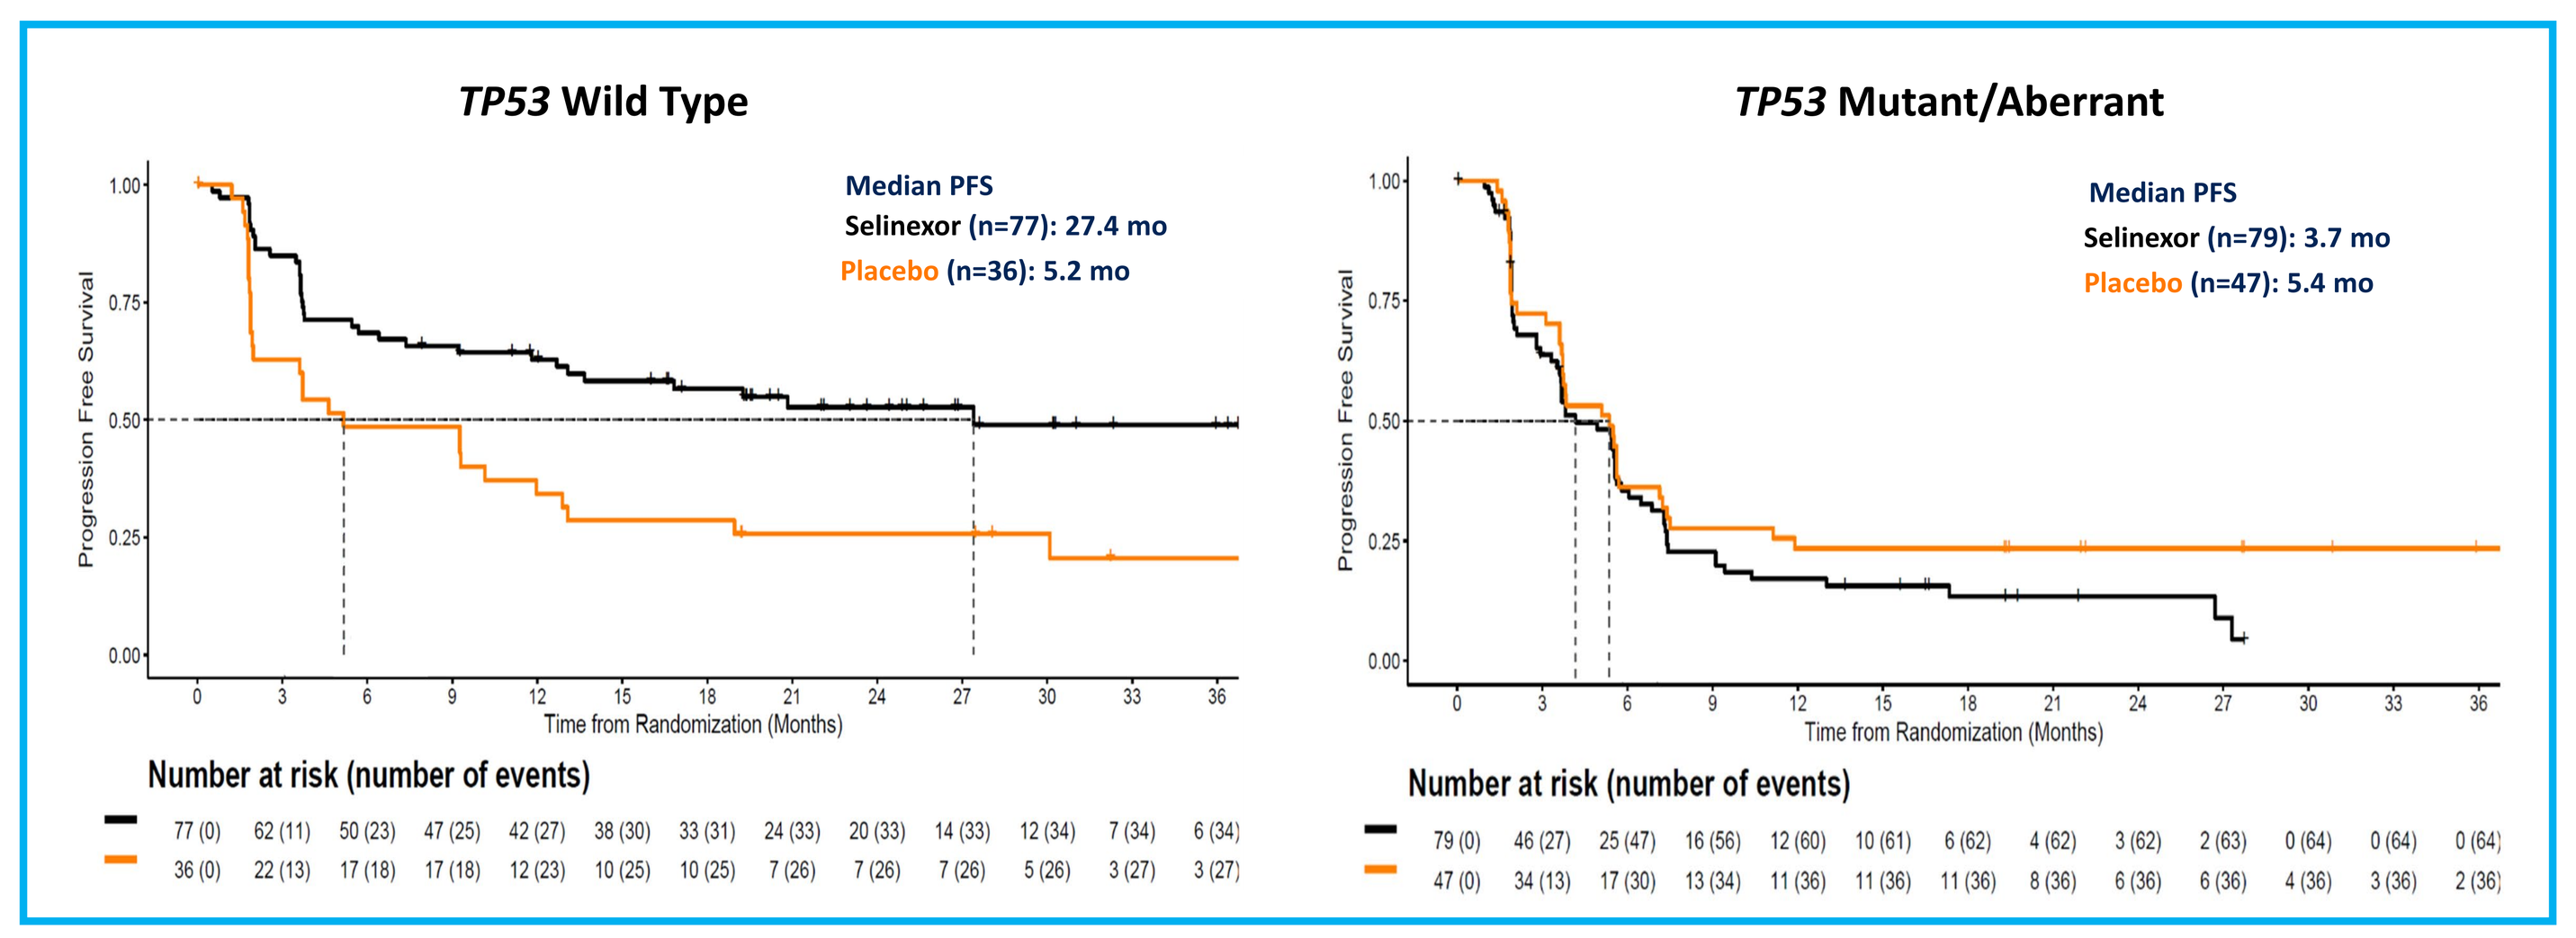
TP53 Wild Type
Median PFS
Selinexor (n=77): 27.4 mo
Placebo (n=36): 5.2 mo
Progression Free Survival
Time from Randomization (Months)
Number at risk (number of events)
77 (0) 62 (11) 50 (23) 47 (25) 42 (27) 38 (30) 33 (31) 24 (33) 20 (33) 14 (33) 12 (34) 7 (34) 6 (34)
36 (0) 22 (13) 17 (18) 17 (18) 12 (23) 10 (25) 10 (25) 7 (26) 7 (26) 7 (26) 5 (26) 3 (27) 3 (27)
TP53 Mutant/Aberrant
Median PFS
Selinexor (n=79): 3.7 mo
Placebo (n=47): 5.4 mo
Progression Free Survival
Time from Randomization (Months)
Number at risk (number of events)
79 (0) 46 (27) 25 (47) 16 (56) 12 (60) 10 (61) 6 (62) 4 (62) 3 (62) 2 (63) 0 (64) 0 (64) 0 (64)
47 (0) 34 (13) 17 (30) 13 (34) 11 (36) 11 (36) 11 (36) 8 (36) 6 (36) 6 (36) 4 (36) 3 (36) 2 (36)

Figure 2. Patients with *TP53* wild-type EC showed longer progression-free survival in the selinexor arm compared to placebo (left), with no benefit in patients with *TP53* mutant EC (right).

- As XPO1 inhibition drives nuclear retention and functional activation of tumor suppressor proteins like p53, we investigated whether *TP53* wild type (WT) status could serve as a predictor for cancer cell sensitivity to XPO1 inhibitors.

METHODS

- Patient derived organoid (PDO) and xenograft (PDX) models were screened for *TP53* gene mutations. Models representing 18 cancer types were included. Within each cancer type, biological replicate models with *TP53* WT and biological replicate models with*TP53* mutations were selected for comparison. For *TP53* mutant models, we focused on testing point mutations within the DNA binding domain (AA95-292) that produced an amino acid change.[2]
- SEL, ELT and control agent (staurosporine for PDO or cisplatin for PDX) half maximal inhibitory concentrations (IC_{50}) after 5 days of exposure were calculated using CellTiter Glo assay. Sensitivity to SEL and ELT was considered *TP53* WT dependent for each cancer type if the IC_{50} for the *TP53* mutant models were ≥1.5-fold higher than the average IC_{50} for the *TP53* WT models.
- IC_{50} of SEL and ELT at 72h was determined for *TP53* WT HCT116 (colorectal) and SNG-M (endometrial) cell lines with syngeneic *TP53* mutant lines[3,4] using CellTiter Glo assays at 72h.
- Quantitative spatial proteomics was conducted in HCT116 cells using isobaric labelling to compare protein expressions in the nucleus of SEL treated vs. control cells, including those with *TP53* WT, *TP53* R175H and *TP53* R273H. Cells were exposed to SEL IC_{50} for 6h to induce proteomic changes without significant cell death. Protein pathway membership was assigned using WikiPathways.

RESULTS

Table 1. Cancer types with *TP53* WT-dependent XPO1 inhibitor sensitivity

Cancer Type	*TP53* Status	Selinexor IC_{50} µM	Fold Δ	Eltanexor IC_{50} µM	Fold Δ	Control IC_{50} µM
Endometrial	WT	0.061		0.067		4.042
	WT	0.031		0.019		0.933
	p.Y220C	0.333	7	0.147	3.4	2.813
	p.R213Ter	1.542	33	1.955	45	6.502
Ovarian	WT	0.089		0.3256		0.0161
	WT	0.051		0.022		0.005
	p. R273C	0.419	6	0.372	2	0.0008
	p. R175H	0.964	13.8	1.215	7	0.0336
Kidney	WT	0.0261		0.027		4.243
	WT	0.074		0.045		8.63
	p.Q167Ter	11.932	238	4.298	118	9.640
	p.R273H	0.101	2	0.091	2.5	3.936
Hepato-cellular	WT	0.119		0.057		0.0122
	WT	0.602		0.514		0.0197
	p.E271Ter	2.601	7	2.845	10	0.0062
	p.R175H	3.855	10.7	4.408	15	0.5833
Esophageal	WT	0.225		0.083		0.0054
	p.R273H	0.391	1.7	0.547	6.5	0.0289
	WT	0.083		0.073		0.9661
	p.R273H	1.448	17.4	1.098	15	6.8916
NSCLC Adeno-carcinoma	WT	0.334		1.855		0.0096
	WT	0.129		0.114		0.0009
	p.F341C	3.455	15	7.516	7.6	0.002
	p.G245S	25.399	110	13.464	13.7	0.008

Ter=Termination PDX Control Cisplatin= Blue
PDO Control Staurosporine=Green

Table 2. Cancer types with *TP53* WT-independent XPO1 inhibitor sensitivity

Cancer Type	*TP53* Status	Selinexor IC_{50} µM	Fold Δ	Eltanexor IC_{50} µM	Fold Δ	Control IC_{50} µM
Head & Neck	WT	0.124		0.129		0.0008
	p.R273H	0.368	3	0.142	1.1	0.0038
	WT	0.089		0.064		10.591
	p.R175H	0.108	1.2	0.094	1.5	1.384
Gastric	WT	1.324		2.272		0.028
	p.R248W	0.221	0.17	0.108	0.5	0.0007
	WT	0.056		0.076		3.9754
	p.R273H	0.074	1.3	0.052	0.7	1.4319
Gallbladder	WT	0.149		0.098		3.4348
	WT	0.252		0.189		8.0681
	p.R248Q	0.124	0.6	0.176	1.2	3.6319
	p.R175H	0.181	0.9	0.159	1.1	4.5145
Prostate Adeno-carcinoma	WT	0.086		0.119		9.775
	WT	0.152		0.133		7.879
	p.Y163C	0.064	0.5	0.043	0.3	9.544
	p.R280Ter	0.102	0.8	0.108	0.8	2.048
Cholangio-carcinoma	WT	0.509		0.518		0.0063
	WT	0.278		0.719		0.0232
	p.R273C	0.171	0.4	0.162	0.3	0.0032
	p.R248Q	0.371	0.9	0.242	0.4	0.0007
Cervical	WT	0.487		0.814		0.0031
	p.E285K	0.356	0.7	0.3996	0.5	0.0023
	WT	0.167		0.1484		2.37
	p.R175H	0.068	0.4	0.0442	0.3	1.53
Breast ER+/PR+	WT	29.013		24.245		0.151
	WT	0.5891		0.3201		0.0372
	p.R273H	1.0254	0.1	1.6362	0.1	0.0306
	p.H179R	0.383	0.03	0.2362	0.02	0.0452
Mixed Mullerian	WT	0.445		0.440		1.668
	WT	0.105		0.066		3.977
	p.R273H	0.099	0.4	0.073	0.3	0.167
	p.R273C	0.108	0.4	0.073	0.3	5.957
NSCLC Squamous	WT	0.274		0.388		0.0068
	WT	0.401		0.637		0.0019
	p. R273L	0.566	1.7	0.632	1.2	0.0035
	p. Y234H	3.533	10	21.249	41.5	0.0024
Pancreatic	WT	0.223		0.309		0.0037
	WT	0.464		7.178		0.0106
	p. R273H	0.635	1.8	0.738	0.2	0.0031
	p. R273H	0.554	1.6	0.611	0.2	0.0027
Bladder	WT	0.094		0.085		0.0024
	WT	0.263		0.176		0.0144
	p.V272_R273insL	6.116	34	6.209	47	0.0044
	p.W53Ter	0.252	1.4	0.267	2	0.0111
Thyroid	WT	0.262		0.117		3.938
	WT	0.107		0.131		4.709
	p.W146Ter	0.446	2.4	0.440	3.6	6.469
	p.C124Ter	0.063	0.3	0.058	0.5	2.434

- Table 1 shows cancer types where the *TP53* WT models were more sensitive to SEL and ELT than *TP53* mutant models (1.7-100-fold difference in IC_{50}). By contrast, Table 2 shows cancer types where SEL or ELT sensitivity was not associated with *TP53* status.
- *TP53* mutation status conferred resistance to cisplatin in only esophageal models, demonstrating specificity between *TP53* mutation status and XPO1 inhibition sensitivity.
- For *TP53* WT PDX models tested, SEL (IC_{50} 0.026-0.6µM) and ELT (IC_{50} 0.019-0.45µM) reduce viability with an order of magnitude lower dose than the cisplatin (IC_{50} 0.93-10.6µM).
- For PDO models tested, *TP53* WT SEL (IC_{50} 0.051-29µM) and ELT (IC_{50} 0.022-24µM) and *TP53* mutants SEL (IC_{50} 0.17-25.4µM) and ELT (IC_{50} 0.108-21.2µM) had wider sensitivity variations but showed similar overall trends with PDX models.

Figure 3. Introduction of *TP53* mutations conferred resistance to SEL and ELT

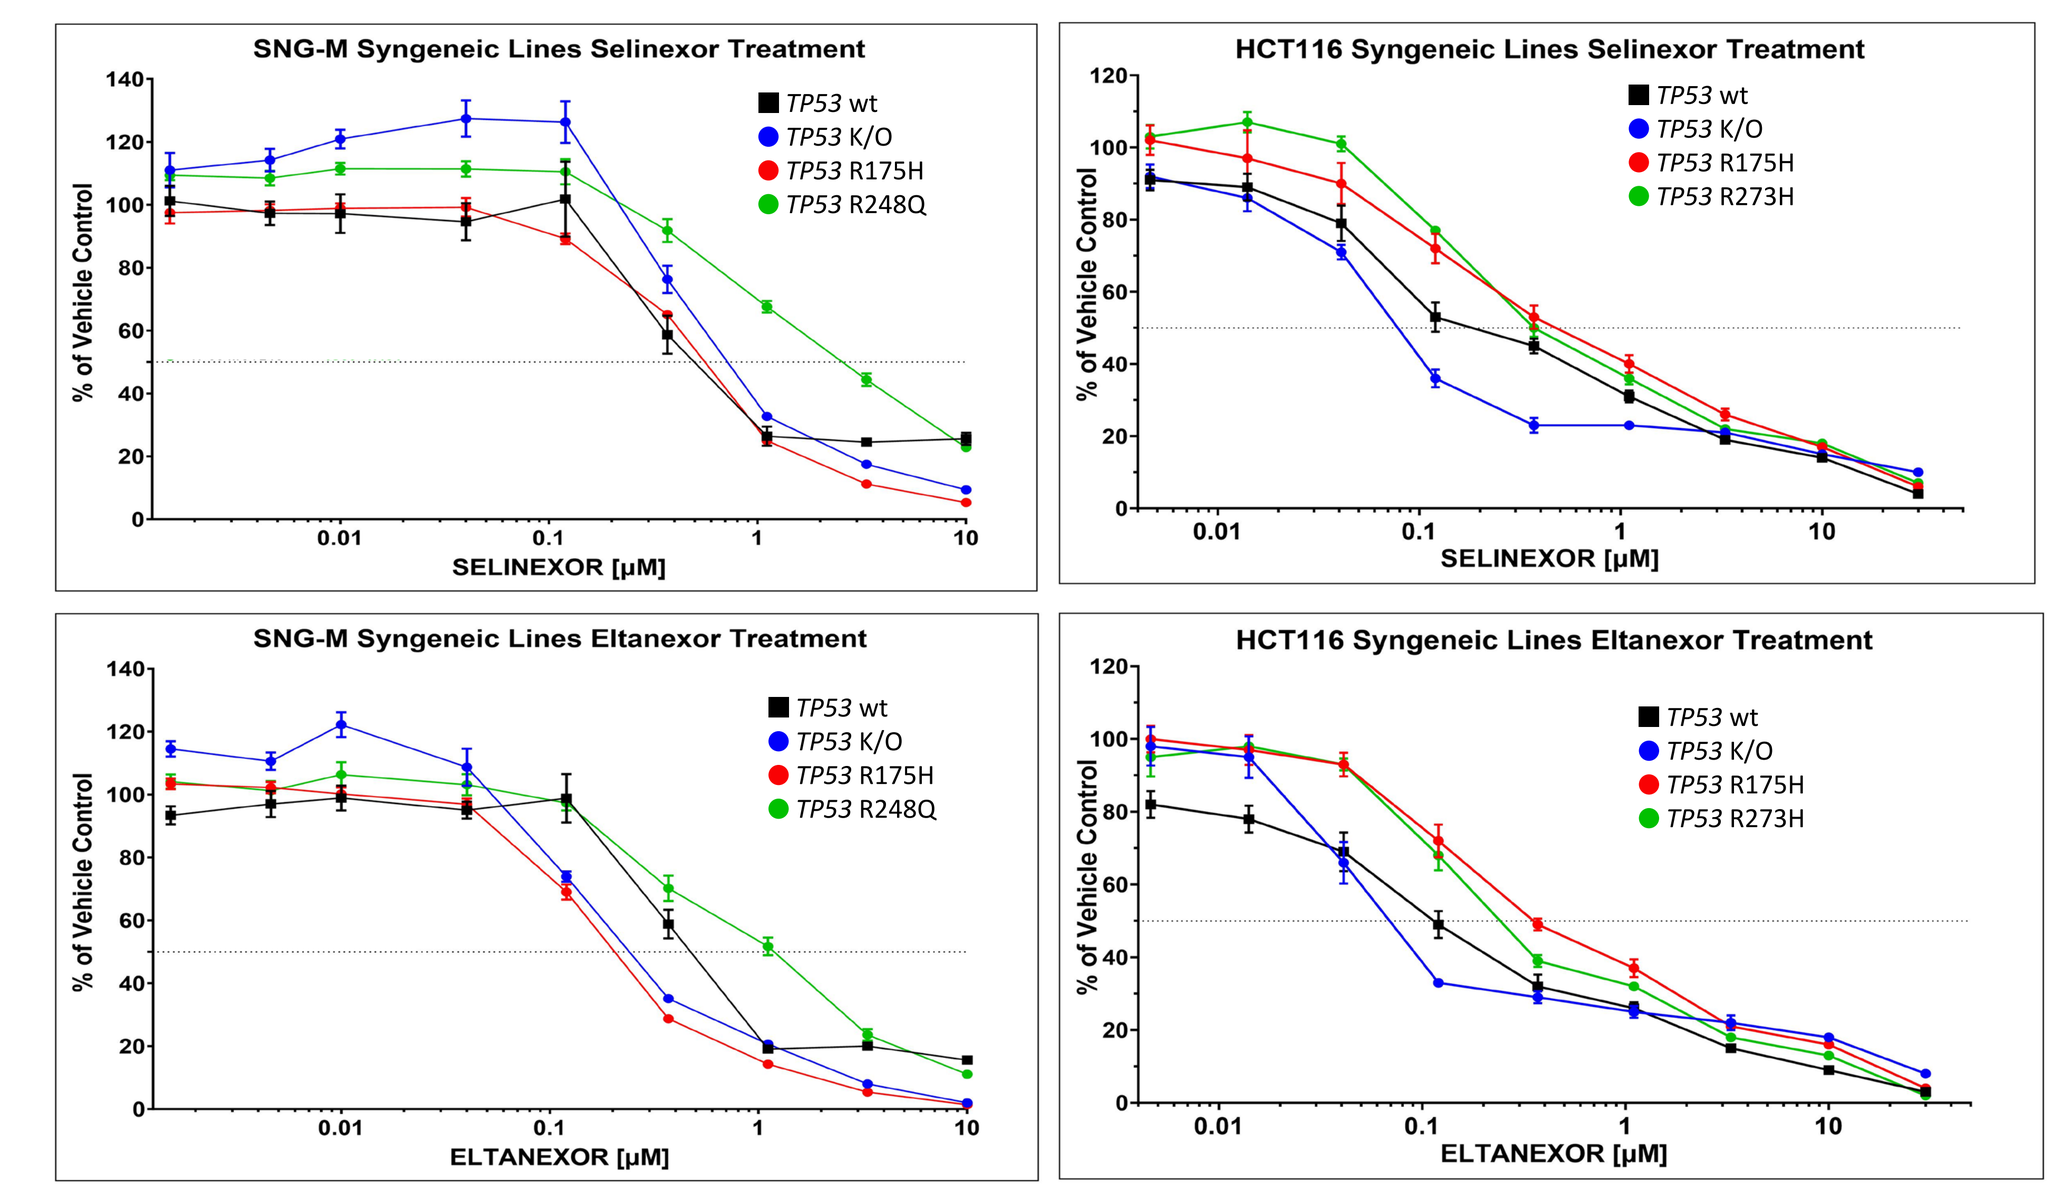
SNG-M Syngeneic Lines Selinexor Treatment
HCT116 Syngeneic Lines Selinexor Treatment
SNG-M Syngeneic Lines Eltanexor Treatment
HCT116 Syngeneic Lines Eltanexor Treatment
TP53 wt
TP53 K/O
TP53 R175H
TP53 R248Q
TP53 R273H
% of Vehicle Control
SELINEXOR [µM]
ELTANEXOR [µM]

SNG-M Cell Lines	Selinexor IC_{50} µM	Fold Δ vs. *P53* WT	Eltanexor IC_{50} µM	Fold Δ vs. *P53* WT
P53 WT NEG CRISPR	0.366	--	0.371	--
P53 KO (Exon 5)	0.487	1.33	0.192	0.52
P53 R175H (Het)	0.511	1.4	0.190	0.51
P53 R248Q (Hom)	1.434	3.92	0.892	2.4

HCT-116 Cell Lines	Selinexor IC_{50} µM	Fold Δ vs. *P53* WT	Eltanexor IC_{50} µM	Fold Δ vs. *P53* WT
P53 WT NEG CRISPR[4]	0.2081	--	0.1736	--
P53 KO (Exon 2)[3]	0.057	0.27	0.0427	0.25
P53 R175H (Hom)[4]	0.343	1.65	0.316	1.83
P53 R273H(Hom)[4]	0.276	1.34	0.240	1.4

- To directly evaluate the association between *TP53* mutation and sensitivity to XPO1 inhibitors, syngeneic EC (SNG-M) and colorectal cancer (HCT-116) cell lines with introduced point mutations or knockouts were used.[3,4] An increase in IC_{50} was observed in cells *TP53* point mutations compared to *TP53* WT (Figure 3).

Figure 4. Quantitative spatial proteomics of SEL treated vs. untreated HCT116 cells with *TP53* WT, *TP53* R175H or *TP53* R273H. (A) Overlap between proteins that were significantly enriched in the nuclear fraction of each cell line after treatment with SEL vs DMSO treated cells. (B-D) Proteins set enrichment analysis of the nuclear enriched proteins. Pathways with proteins enriched (red) or depleted (blue) in the nuclear fractions of SEL treated cells are shown.

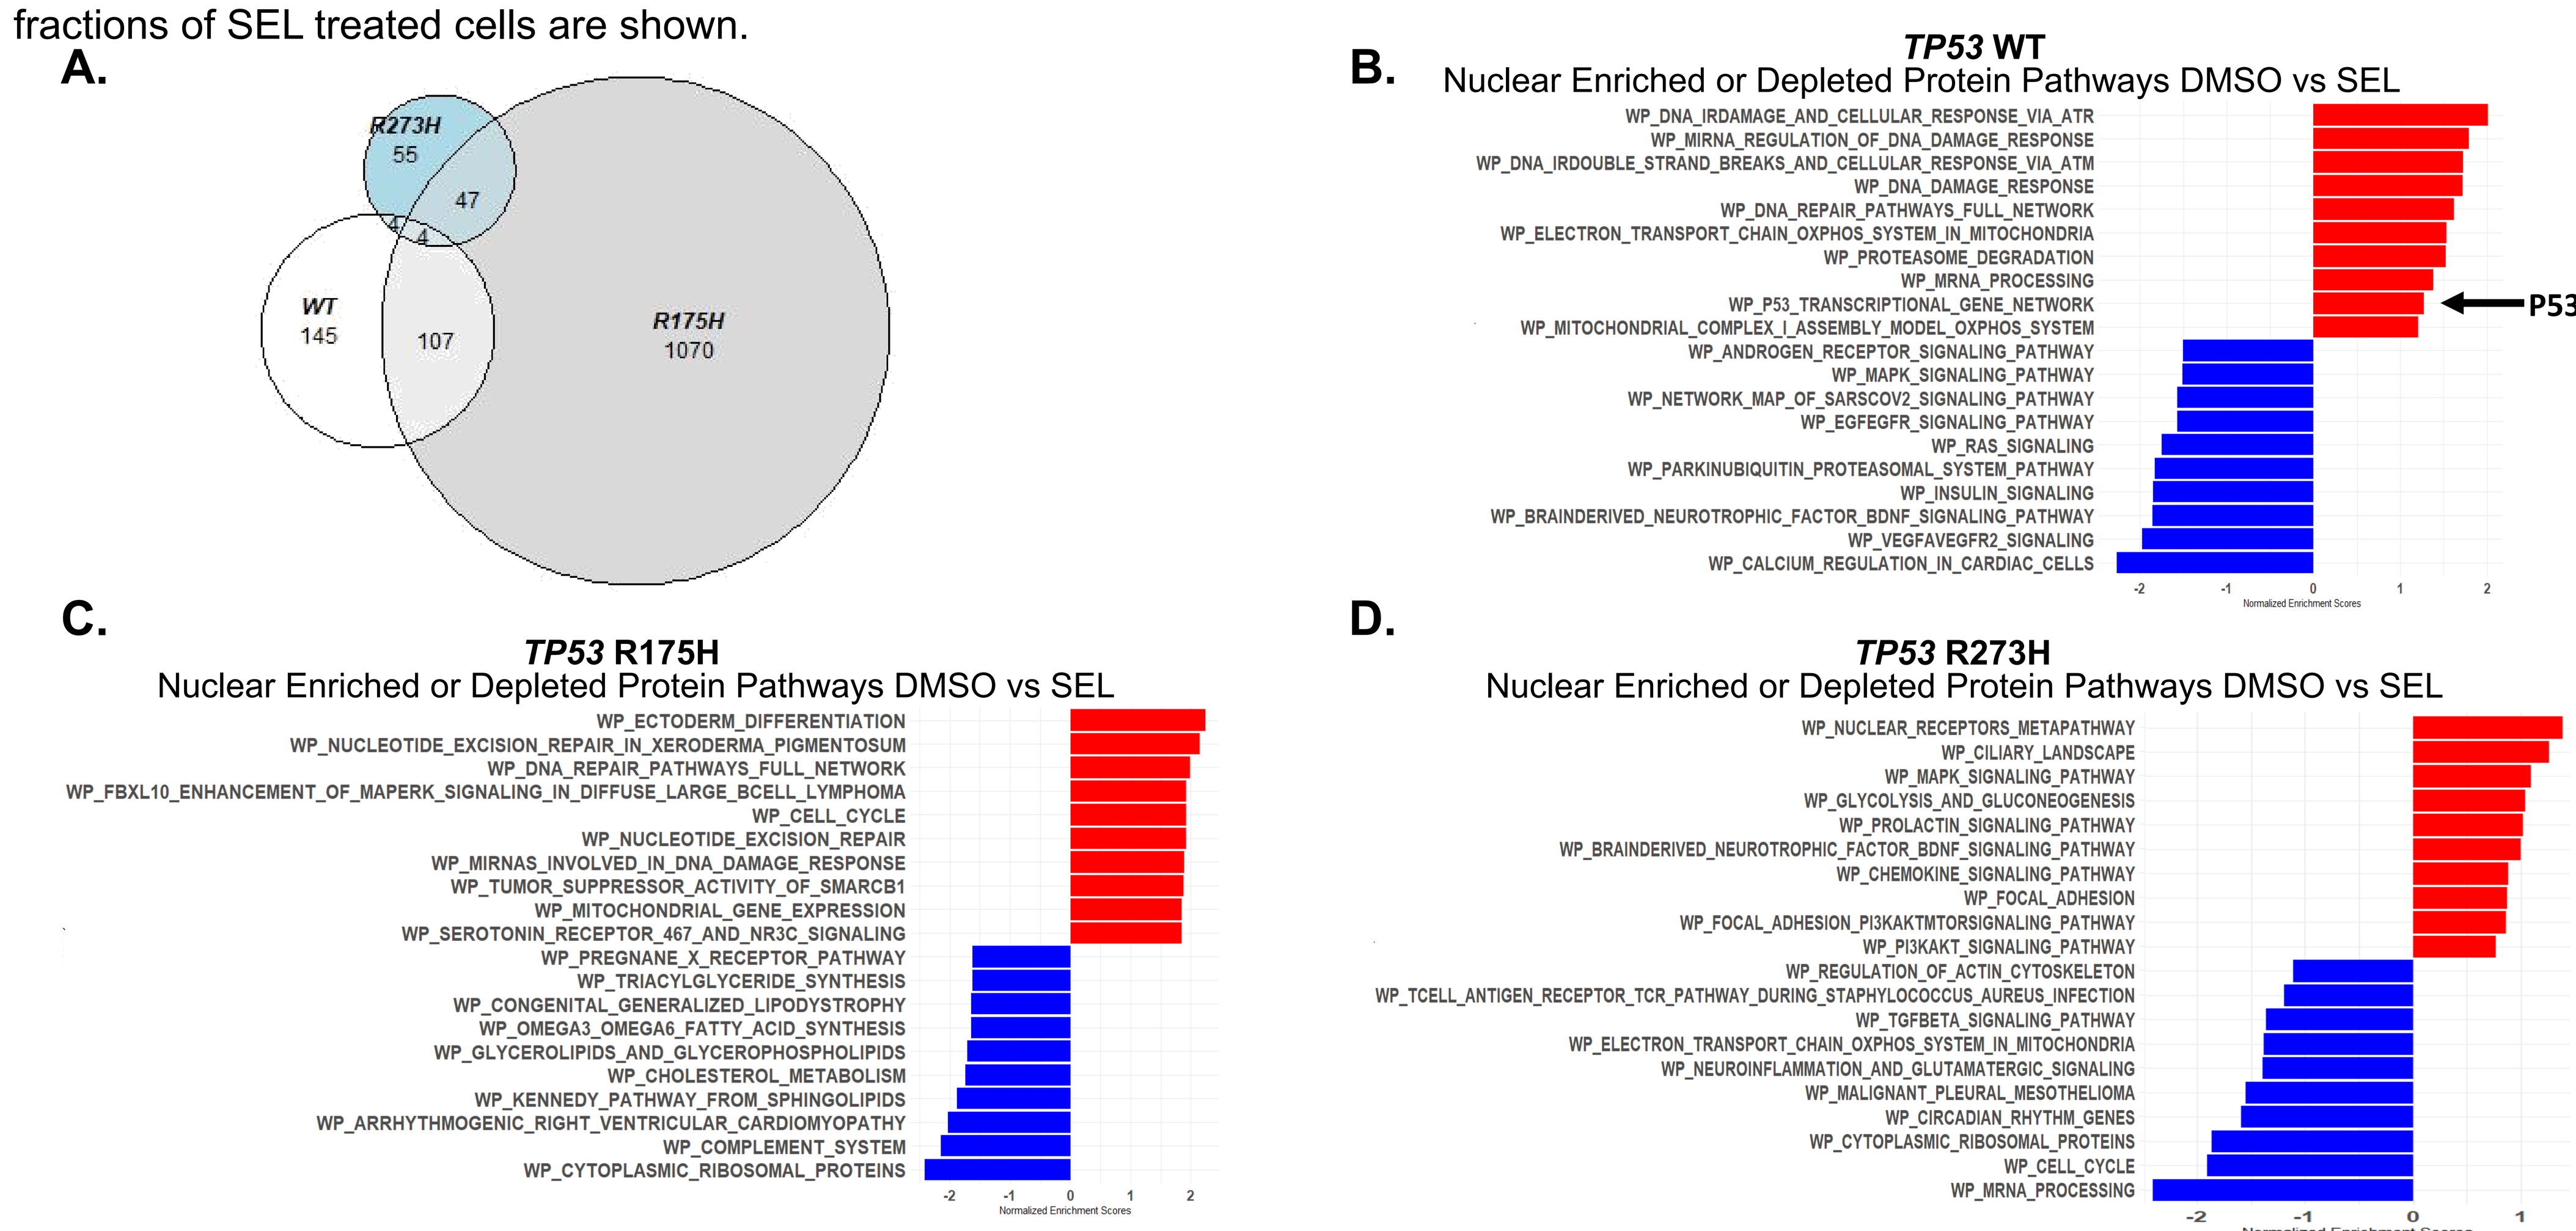
A.
R273H
55
47
4
4
WT
145
107
R175H
1070
B.
TP53 WT
Nuclear Enriched or Depleted Protein Pathways DMSO vs SEL
WP_DNA_IRDAMAGE_AND_CELLULAR_RESPONSE_VIA_ATR
WP_MIRNA_REGULATION_OF_DNA_DAMAGE_RESPONSE
WP_DNA_IRDOUBLE_STRAND_BREAKS_AND_CELLULAR_RESPONSE_VIA_ATM
WP_DNA_DAMAGE_RESPONSE
WP_DNA_REPAIR_PATHWAYS_FULL_NETWORK
WP_ELECTRON_TRANSPORT_CHAIN_OXPHOS_SYSTEM_IN_MITOCHONDRIA
WP_PROTEASOME_DEGRADATION
WP_MRNA_PROCESSING
WP_P53_TRANSCRIPTIONAL_GENE_NETWORK
WP_MITOCHONDRIAL_COMPLEX_I_ASSEMBLY_MODEL_OXPHOS_SYSTEM
WP_ANDROGEN_RECEPTOR_SIGNALING_PATHWAY
WP_MAPK_SIGNALING_PATHWAY
WP_NETWORK_MAP_OF_SARSCOV2_SIGNALING_PATHWAY
WP_EGFEGFR_SIGNALING_PATHWAY
WP_RAS_SIGNALING
WP_PARKINUBIQUITIN_PROTEASOMAL_SYSTEM_PATHWAY
WP_INSULIN_SIGNALING
WP_BRAINDERIVED_NEUROTROPHIC_FACTOR_BDNF_SIGNALING_PATHWAY
WP_VEGFAVEGFR2_SIGNALING
WP_CALCIUM_REGULATION_IN_CARDIAC_CELLS
P53
Normalized Enrichment Scores
C.
TP53 R175H
Nuclear Enriched or Depleted Protein Pathways DMSO vs SEL
WP_ECTODERM_DIFFERENTIATION
WP_NUCLEOTIDE_EXCISION_REPAIR_IN_XERODERMA_PIGMENTOSUM
WP_DNA_REPAIR_PATHWAYS_FULL_NETWORK
WP_FBXL10_ENHANCEMENT_OF_MAPERK_SIGNALING_IN_DIFFUSE_LARGE_BCELL_LYMPHOMA
WP_CELL_CYCLE
WP_NUCLEOTIDE_EXCISION_REPAIR
WP_MIRNAS_INVOLVED_IN_DNA_DAMAGE_RESPONSE
WP_TUMOR_SUPPRESSOR_ACTIVITY_OF_SMARCB1
WP_MITOCHONDRIAL_GENE_EXPRESSION
WP_SEROTONIN_RECEPTOR_467_AND_NR3C_SIGNALING
WP_PREGNANE_X_RECEPTOR_PATHWAY
WP_TRIACYLGLYCERIDE_SYNTHESIS
WP_CONGENITAL_GENERALIZED_LIPODYSTROPHY
WP_OMEGA3_OMEGA6_FATTY_ACID_SYNTHESIS
WP_GLYCEROLIPIDS_AND_GLYCEROPHOSPHOLIPIDS
WP_CHOLESTEROL_METABOLISM
WP_KENNEDY_PATHWAY_FROM_SPHINGOLIPIDS
WP_ARRHYTHMOGENIC_RIGHT_VENTRICULAR_CARDIOMYOPATHY
WP_COMPLEMENT_SYSTEM
WP_CYTOPLASMIC_RIBOSOMAL_PROTEINS
Normalized Enrichment Scores
D.
TP53 R273H
Nuclear Enriched or Depleted Protein Pathways DMSO vs SEL
WP_NUCLEAR_RECEPTORS_METAPATHWAY
WP_CILIARY_LANDSCAPE
WP_MAPK_SIGNALING_PATHWAY
WP_GLYCOLYSIS_AND_GLUCONEOGENESIS
WP_PROLACTIN_SIGNALING_PATHWAY
WP_BRAINDERIVED_NEUROTROPHIC_FACTOR_BDNF_SIGNALING_PATHWAY
WP_CHEMOKINE_SIGNALING_PATHWAY
WP_FOCAL_ADHESION
WP_FOCAL_ADHESION_PI3KAKTMTORSIGNALING_PATHWAY
WP_PI3KAKT_SIGNALING_PATHWAY
WP_REGULATION_OF_ACTIN_CYTOSKELETON
WP_TCELL_ANTIGEN_RECEPTOR_TCR_PATHWAY_DURING_STAPHYLOCOCCUS_AUREUS_INFECTION
WP_TGFBETA_SIGNALING_PATHWAY
WP_ELECTRON_TRANSPORT_CHAIN_OXPHOS_SYSTEM_IN_MITOCHONDRIA
WP_NEUROINFLAMMATION_AND_GLUTAMATERGIC_SIGNALING
WP_MALIGNANT_PLEURAL_MESOTHELIOMA
WP_CIRCADIAN_RHYTHM_GENES
WP_CYTOPLASMIC_RIBOSOMAL_PROTEINS
WP_CELL_CYCLE
WP_MRNA_PROCESSING
Normalized Enrichment Scores

- Spatial proteomics quantified 6,893 proteins in nuclear fractions of HCT-116 cells treated with equi-toxic concentrations of SEL. Among the different cell lines, SEL treatment enhanced the nuclear expression of 260 proteins (*TP53* WT), 1,229 proteins (*TP53* R175H) and 110 proteins (*TP53* R273H).
- Minimal overlap in SEL-induced nuclear retained proteins was seen between the cell lines, consistent with context-specific mechanism of action of SEL. (Figure 4A).
- Pathway analysis was performed on nuclear retained proteins in the different cell lines. P53 pathway proteins were only significantly enriched in the nuclear fraction of the *TP53* WT cells. Other pathway differences were evident (Figure 4B-D).
- Some proteins were depleted in the nuclei of SEL treated cells compared to controls, indicating indirect effects of XPO1 inhibition, as XPO1 exclusively exports proteins from the nucleus to the cytoplasm.

CONCLUSIONS

- ***TP53* WT status can be a biomarker of sensitivity to the XPO1 inhibitors SEL and ELT in multiple cancer types tested, including endometrial (~20 fold more sensitive), ovarian, kidney, liver, esophageal, lung, pancreatic, and bladder. *TP53* WT models within these cancer types show similar sensitivity to SEL and ELT.**
- ***TP53* WT cell lines were more sensitive to SEL and ELT compared to the same lines with CRISPR engineered *TP53* point mutations.**
- **Spatial proteomics revealed differences in SEL-induced nuclear protein retention between *TP53* mutant and WT cell lines treated with equitoxic concentrations. In *TP53* WT cells, enriched nuclear proteins were members of canonical tumor suppressor pathways including the *TP53* network. In *TP53* mutant lines, proteins were members of DNA-damage, metabolic and cell cycle arrest pathways.**

REFERENCES: 1. Slomovitz B. Presentation presented at: ASCO Plenary Series; 2023 Jul 25; Virtual. 2. Joerger AC et al. *Annu Rev Biochem*. 2016 Jun 2;85:375–404. 3. Bunz F et al. *Science*. 1998 Nov 20;282(5393):1497–501. 4. Hassin O. et al. *Nat Commun*. 2022 May 19;13(1):2800.

ACKNOWLEDGEMENTS: The authors thank Steve Mitchell, Trinayan Kashyap, and Liwen Zhang for editorial and scientific assistance with this work.

AACR-NCI-EORTC International Conference on Molecular Targets and Cancer Therapeutics
Oct 11-15, 2023 Boston, MA

Karyopharm Therapeutics